AK112, a tetravalent bispecific antibody targeting PD-1 and VEGF, enhances binding avidity and functional activities and elicits potent anti-tumor efficacy in pre-clinical studies

No.521

Tingting Zhong, Zhaoliang Huang, Xinghua Pang, Chunshan Jin, Xinrong He, Michelle Xia, Baiyong Li, Jing Min.
Akeso Biopharma Co., Ltd., Zhongshan, Guangdong Province, China. For correspondence, please contact: jing.min@akesobio.com

Introduction

PD-1/PD-L1 inhibition immunotherapy holds great promise in cancer treatment. Combination treatment using anti-PD-1/PD-L1 agents with other immunotherapeutics brings additional benefits, such as preventing refractory effects towards PD-1/PD-L1 antibodies, and improving anti-tumor activities. Vascular endothelial growth factor (VEGF) is found to be frequently overexpressed in various solid tumors, which not only promotes tumor angiogenesis but also functions to suppress anti-tumor immune response [1, 2]. Consequently, a novel anti-PD-1/VEGF bispecific antibody (AK112) was designed to inhibit PD-1-mediated immunosuppression and simultaneously block tumor angiogenesis in the tumor microenvironment (TME). The tetravalent structure of AK112 allows formation of large complexes with dimeric VEGF, resulting in improved avidity to PD-1 and functional activities, which elicits potent anti-tumor efficacy in pre-clinical studies.

Figure 1. Schematic diagram of the structure of AK112 and mechanism of action

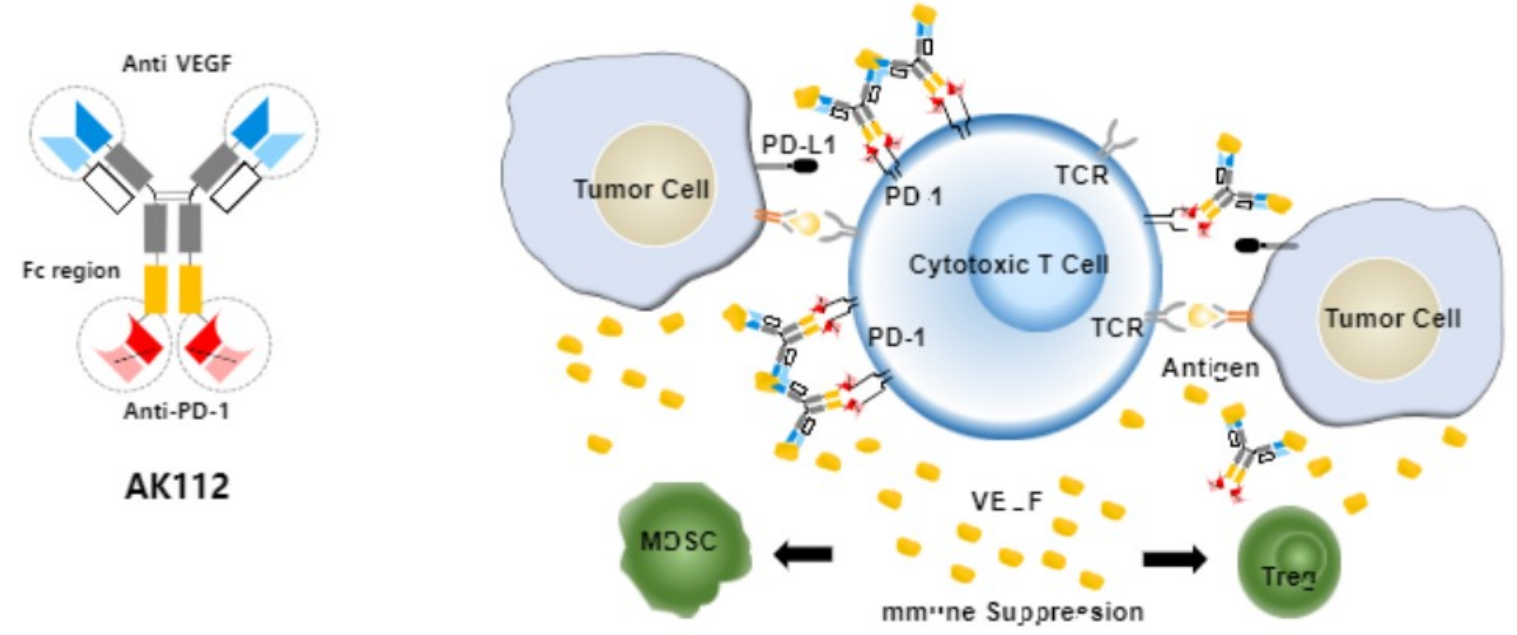
Anti VEGF
Fc region
Anti-PD-1
AK112
Tumor Cell
PD-L1
PD-1
TCR
Cytotoxic T Cell
TCR
PD-1
Antigen
Tumor Cell
VEGF
MDSC
Treg
Immune Suppression

Methods

The antigen binding activity of AK112 with PD-1 and VEGF were assessed by ELISA, Fortebio and flow cytometry. The formation of AK112-VEGF complexes was detected by size-exclusion high-pressure liquid chromatography (SEC-HPLC). To determine if VEGF could enhance the avidity of AK112 to PD-1, the binding activity of AK112 with PD-1 was evaluated by Fortebio and flow cytometry in the presence of VEGF. The blockade of PD-1/PD-L1 signaling pathway was determined in luciferase reporter cell assay. The PD-1 internalization was determined by flow cytometry. In in-vivo pharmacology studies, the anti-tumor activity of AK112 was investigated in SCID/Beige mice implanted with HCC827 cells.

Results

Table 1 Antigen binding activity of AK112 to PD-1 and VEGFA

Antibody	EC_{50} (nM) of antigen binding activity			K_D (M) of antigen binding affinity	
	ELISA[a]		FACS[b]	Fortebio[c]	
	PD-1	VEGFA	293T-PD-1 cells	PD-1	VEGFA
AK112	0.060	0.036	3.49	2.4[illegible]E-10	3.30E-10
Bevacizumab	NA	0.035	NA	NA	6.04E-10
Nivolumab	0.044	NA	2.10	2.48E-10	NA

a, fusion protein of mouse Fc with human PD-1 extracellular domain or 6× histidine-tagged VEGFA protein was fixed onto the plates in the assays; b, 293T cells transfected with human PD-1 (293T-PD-1 cells) were used as target cells in the assay; c, biotinylated fusion protein of human Fc with human PD-1 extracellular domain or 6× histidine-tagged VEGFA protein was immobilized onto the sensor in the assays. NA, not applicable.

Results

Figure 2. AK112-VEGFA complex formation determined by SEC-HPLC.

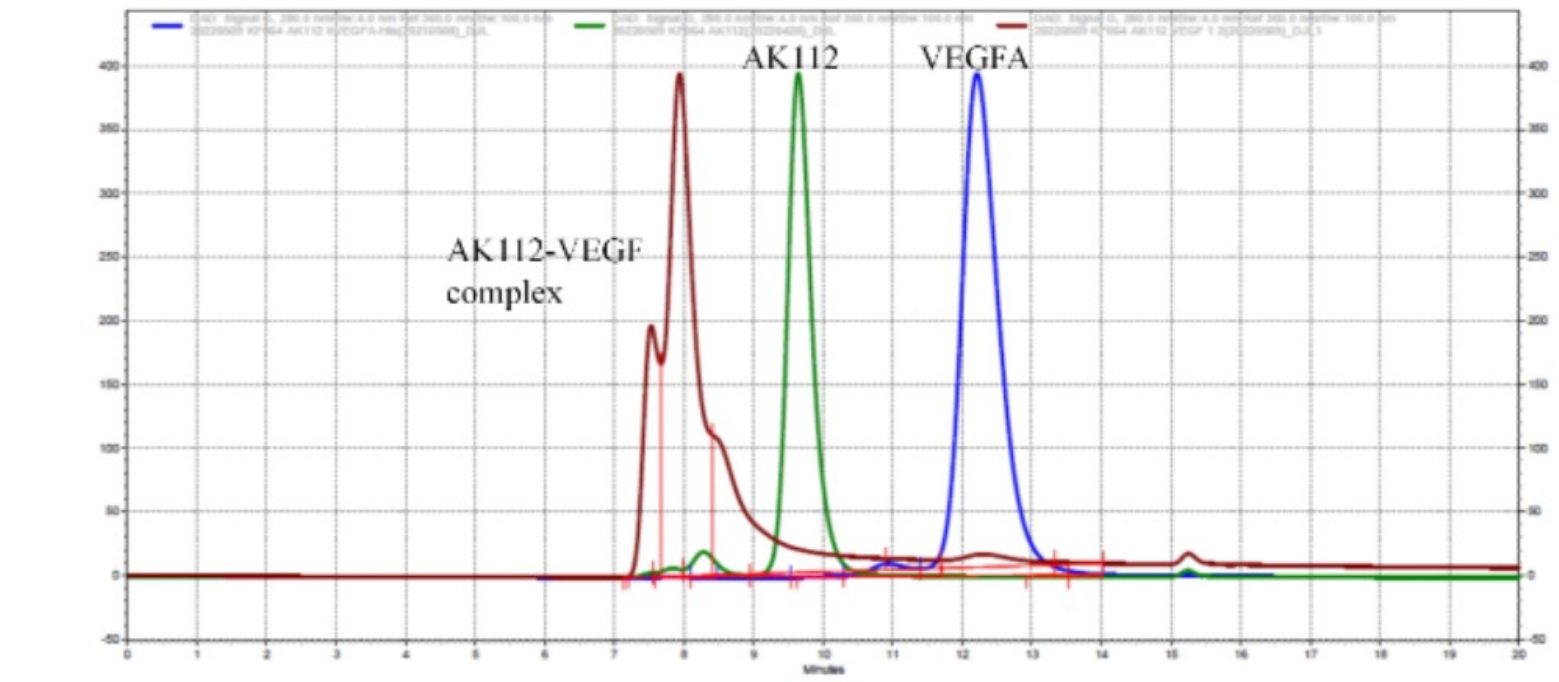
AK112
VEGFA
AK112-VEGF complex
Minutes

AK112 were premixed with 2× VEGFA and then analyzed on SEC-HPLC (Red color). AK112 alone (Green color) and VEGFA alone (Blue color) were included as reference.

Table 2. Promoted binding avidity of AK112 to human PD-1 in the presence of VEGFA.

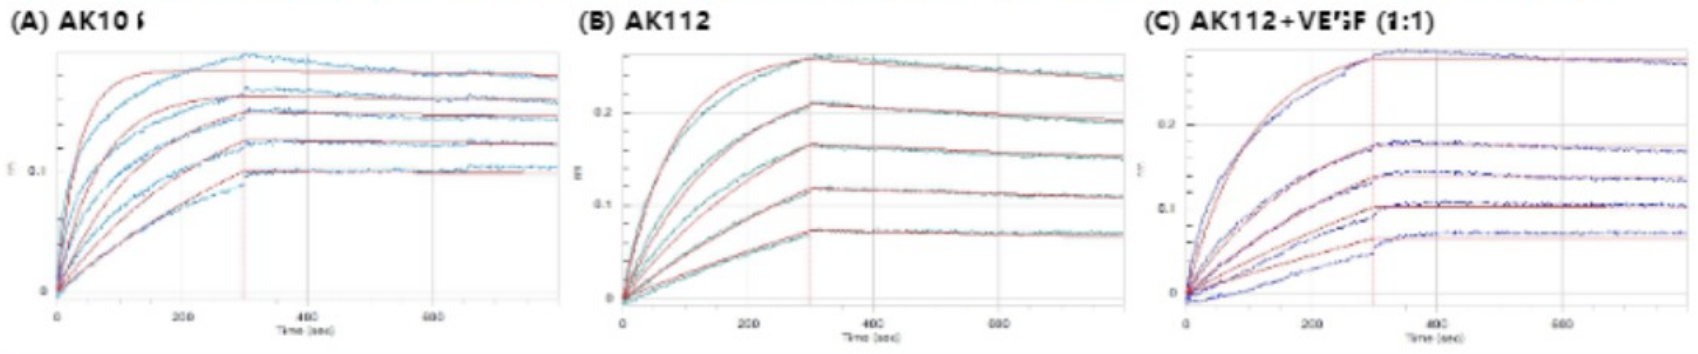
(A) AK105
(B) AK112
(C) AK112+VEGF (1:1)

Fixed antigen	Antibody[a]	VEGF-A-his (nM)	KD (M)	kon (1/ms)	kdis (1/s)
PD1-his 200 nM	AK105	0	4.11E-10	5.74E+05	2.36E-04
	AK112	0	7.15E-10	2.94E+05	2.10E-04
	AK112+VEGF[b]	50-1.56	3.83E-11	2.51E+05	9.62E-06

a, antibodies with two-fold serial dilution from 50 nM to 1.56 nM; b, AK112 was pre-incubated with human VEGFA-his at same concentration and then diluted from 50 nM to 1.56 nM.

Figure 3. Antigen binding activity of AK112 to PD-1 expressing cells.

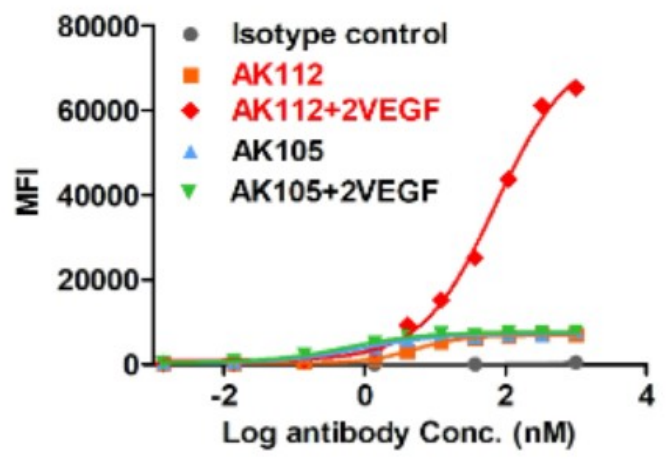
80000
60000
40000
20000
0
MFI
-2
0
2
4
Log antibody Conc. (nM)
Isotype control
AK112
AK112+2VEGF
AK105
AK105+2VEGF

FACS binding curves of AK112 and AK105 with or without 2×VEGFA to PD-1 on PD-1 transfected Jurkat cells. Secondary antibody is mouse anti-Human IgG FC-Alexa Fluuor 647. MFI, mean fluorescent intensity.

Figure 4. VEGFA effect on bioactivity of AK112 to enhance PD-1 internalization.

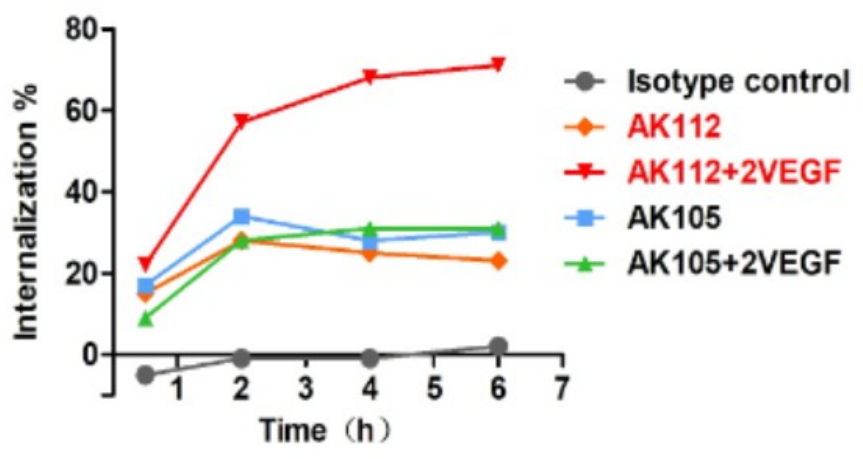
80
60
40
20
0
Internalization %
1
2
3
4
5
6
7
Time (h)
Isotype control
AK112
AK112+2VEGF
AK105
AK105+2VEGF

Cell surface PD-1 level on PD-1-expressing Jurkat cells were detected by FACS at different time points after AK112 and AK105 treatment with or without VEGFA. Internalization percentage was calculated from the decrease of surface PD-1 compared to its expression at 0 h.

Results

Figure 5. Enhanced bioactivity of AK112 to block PD-1/PD-L1 signaling pathway in the presence of VEGFA.

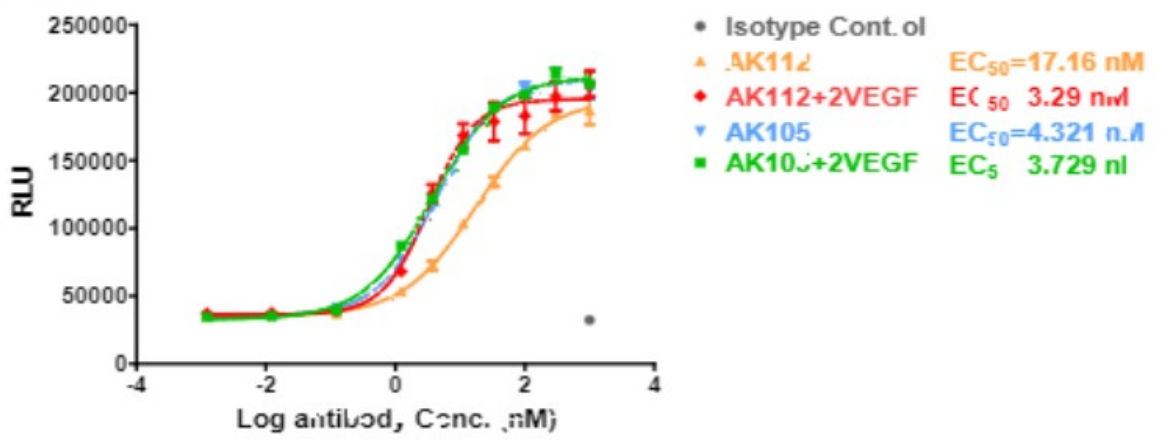
250000
200000
150000
100000
50000
0
RLU
-4
-2
0
2
4
Log antibody Conc. (nM)
Isotype Control
AK112 EC50=17.16 nM
AK112+2VEGF EC50=3.29 nM
AK105 EC50=4.321 nM
AK105+2VEGF EC50=3.729 nM

AK112 and anti-PD-1 antibody AK105 with or without 2×VEGFA blocked the interaction of PD-1 and PD-L1, leading to enhancement of luminescence in the co-culture of PD-L1 aAPC/CHO-K1 cells and PD-1 effector cells. Luminescence signals were detected by Steady-Glo Luciferase assay. RLU, relative light units.

Figure 6. AK112 inhibited tumor growth in SCID/Beige mice with subcutaneous HCC827 tumor.

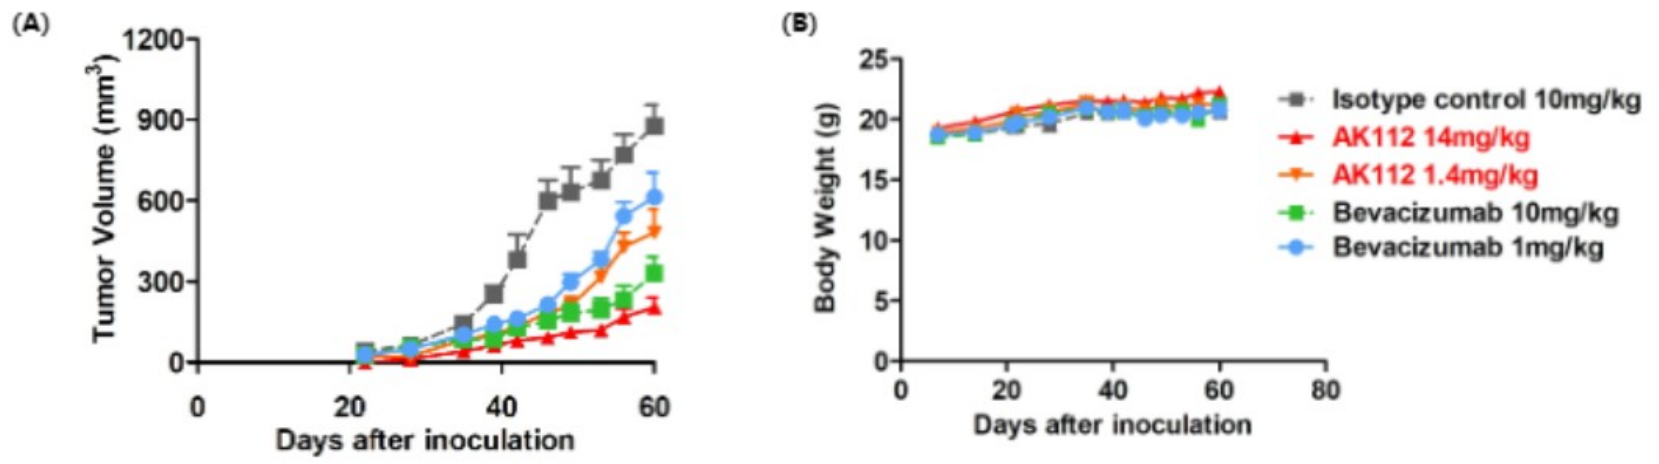
(A)
1200
900
600
300
0
Tumor Volume (mm3)
0
20
40
60
Days after inoculation
(B)
25
20
15
10
5
0
Body Weight (g)
0
20
40
60
80
Days after inoculation
Isotype control 10mg/kg
AK112 14mg/kg
AK112 1.4mg/kg
Bevacizumab 10mg/kg
Bevacizumab 1mg/kg

Each mouse was inoculated subcutaneously at the right hind flank with HCC827 cells, PBMCs and AK112, Bevacizumab or isotype control anti-HEL mixture on day 0. Different doses of antibodies were then continuously intravenously injected on day 7, 14, 21, 28, 35. (A) Tumor volume and (B) body weight were measured.

Conclusion

AK112 could specifically bind to human PD-1 and VEGF with high affinity (Table 1). Intriguingly, AK112 was found to form soluble complexes with VEGF by SEC-HPLC assay (Figure 2). Notably, VEGF efficiently enhanced the binding of AK112 to PD-1 (Table 2, Figure 3), which led to increased PD-1 internalization (Figure 4) and better potency on blockade of PD-1/PD-L1 signaling (Figure 5) relevant to AK112 alone. Moreover, AK112 demonstrated greater anti-tumor efficacy compared to Bevacizumab in mice (Figure 6).

AK112, a dual-blocking anti-PD-1/VEGF bispecific antibody, shows improved avidity to PD-1 in the presence of VEGF, and displays great anti-tumor efficacy in a mouse tumor model, supporting its clinical development for the treatment of human cancers.

Reference

[1] Carmeliet P. VEGF as a key mediator of angiogenesis in cancer. Oncology 2005;69 Suppl 3:4-10. doi: 10.1159/000088478. Epub 2005 Nov 21. PMID: 16301830.

[2] Ohm JE, Carbone DP. VEGF as a mediator of tumor-associated immunodeficiency. Immunol Res. 2001;23(2-3):263-72. doi: 10.1385/IR:23:2-3:263. PMID: 11444391.

The authors gratefully acknowledge Lingzhi Zhang, Wen Yin and Shuyue Huang (all from Akeso Biopharma, Co., Ltd.) for their contributions to this poster.

Akeso Biopharma Co., Ltd. Presented at SITC 2022 annual meeting. All right reserved