876P

Evaluation of the Safety and Efficacy of Ivonescimab in Combination With Ligufalimab as First-Line Treatment for PD-L1–Positive Recurrent/Metastatic Head and Neck Squamous Cell Carcinoma

Xiaozhong Chen,[1] Man Hu,[2] Ting Jin,[1] Kai Teng,[2] Yaqian Han,[3] Yujuan Zhou,[3] Kunyu Yang,[4] Zhanjie Zhang,[4] Jun Wang,[5] Yunjie Cheng,[5] Wenting Li,[6] Zhongmin Maxwell Wang,[6] Baiyong Li,[6] Michelle Xia[6]

[1]Zhejiang Cancer Hospital, Hangzhou, China; [2]Cancer Hospital of Shandong First Medical University (Shandong Cancer Hospital), Jinan, China; [3]The Affiliated Cancer Hospital of Xiangya School of Medicine, Central South University/Hunan Cancer Hospital, Changsha, China; [4]Cancer Center, Union Hospital of Tongji Medical College, Huazhong University of Science and Technology, Wuhan, China; [5]Fourth Affiliated Hospital of Hebei Medical University, Shijiazhuang, China; [6]Akeso Biopharma Inc., Zhongshan, China

BACKGROUND

- Although programmed cell death protein 1 (PD-1) inhibitors combined with chemotherapy are approved as first-line treatment for recurrent/metastatic (R/M) head and neck squamous cell carcinoma (HNSCC), some patients respond poorly to this treatment, warranting the need for new treatment strategies[1-2]
- Cluster of differentiation 47 (CD47) is a macrophage immune checkpoint protein that is overexpressed in HNSCC; overexpression of CD47 may correlate with elevated tumor programmed death-ligand 1 (PD-L1) levels and other markers of immunosuppression[3,4]
- Ivonescimab is a novel, investigational bispecific antibody against PD-1 and vascular endothelial growth factor (VEGF) that has exhibited cooperative binding in vitro and could produce complementary and synergistic antitumor effects through both the PD-1 and the VEGF pathways[5-7]
- Ligufalimab is a novel humanized immunoglobulin G4 monoclonal antibody to target CD47 that was designed to avoid hemagglutination, which adversely affected the safety profile of previous CD47-blocking antibodies[8,9]
 - › Ligufalimab may enhance antitumor activity when combined with ivonescimab, as demonstrated in xenograft models[8]
- Here, we present data on the efficacy and safety of ivonescimab, both with and without ligufalimab, in patients with PD-L1–positive R/M HNSCC who have not received systemic anticancer therapy for recurrent or metastatic disease

OBJECTIVE

- To assess the safety and efficacy of ivonescimab as monotherapy or in combination with ligufalimab in patients 18-75 years of age with PD-L1–positive R/M HNSCC

METHODS

Study Design

- In this open-label, multicenter phase 2 trial (NCT05229497), patients in China with R/M HNSCC were assigned to receive ivonescimab monotherapy or ivonescimab in combination with ligufalimab (**Figure 1**)

Figure 1. Study Design

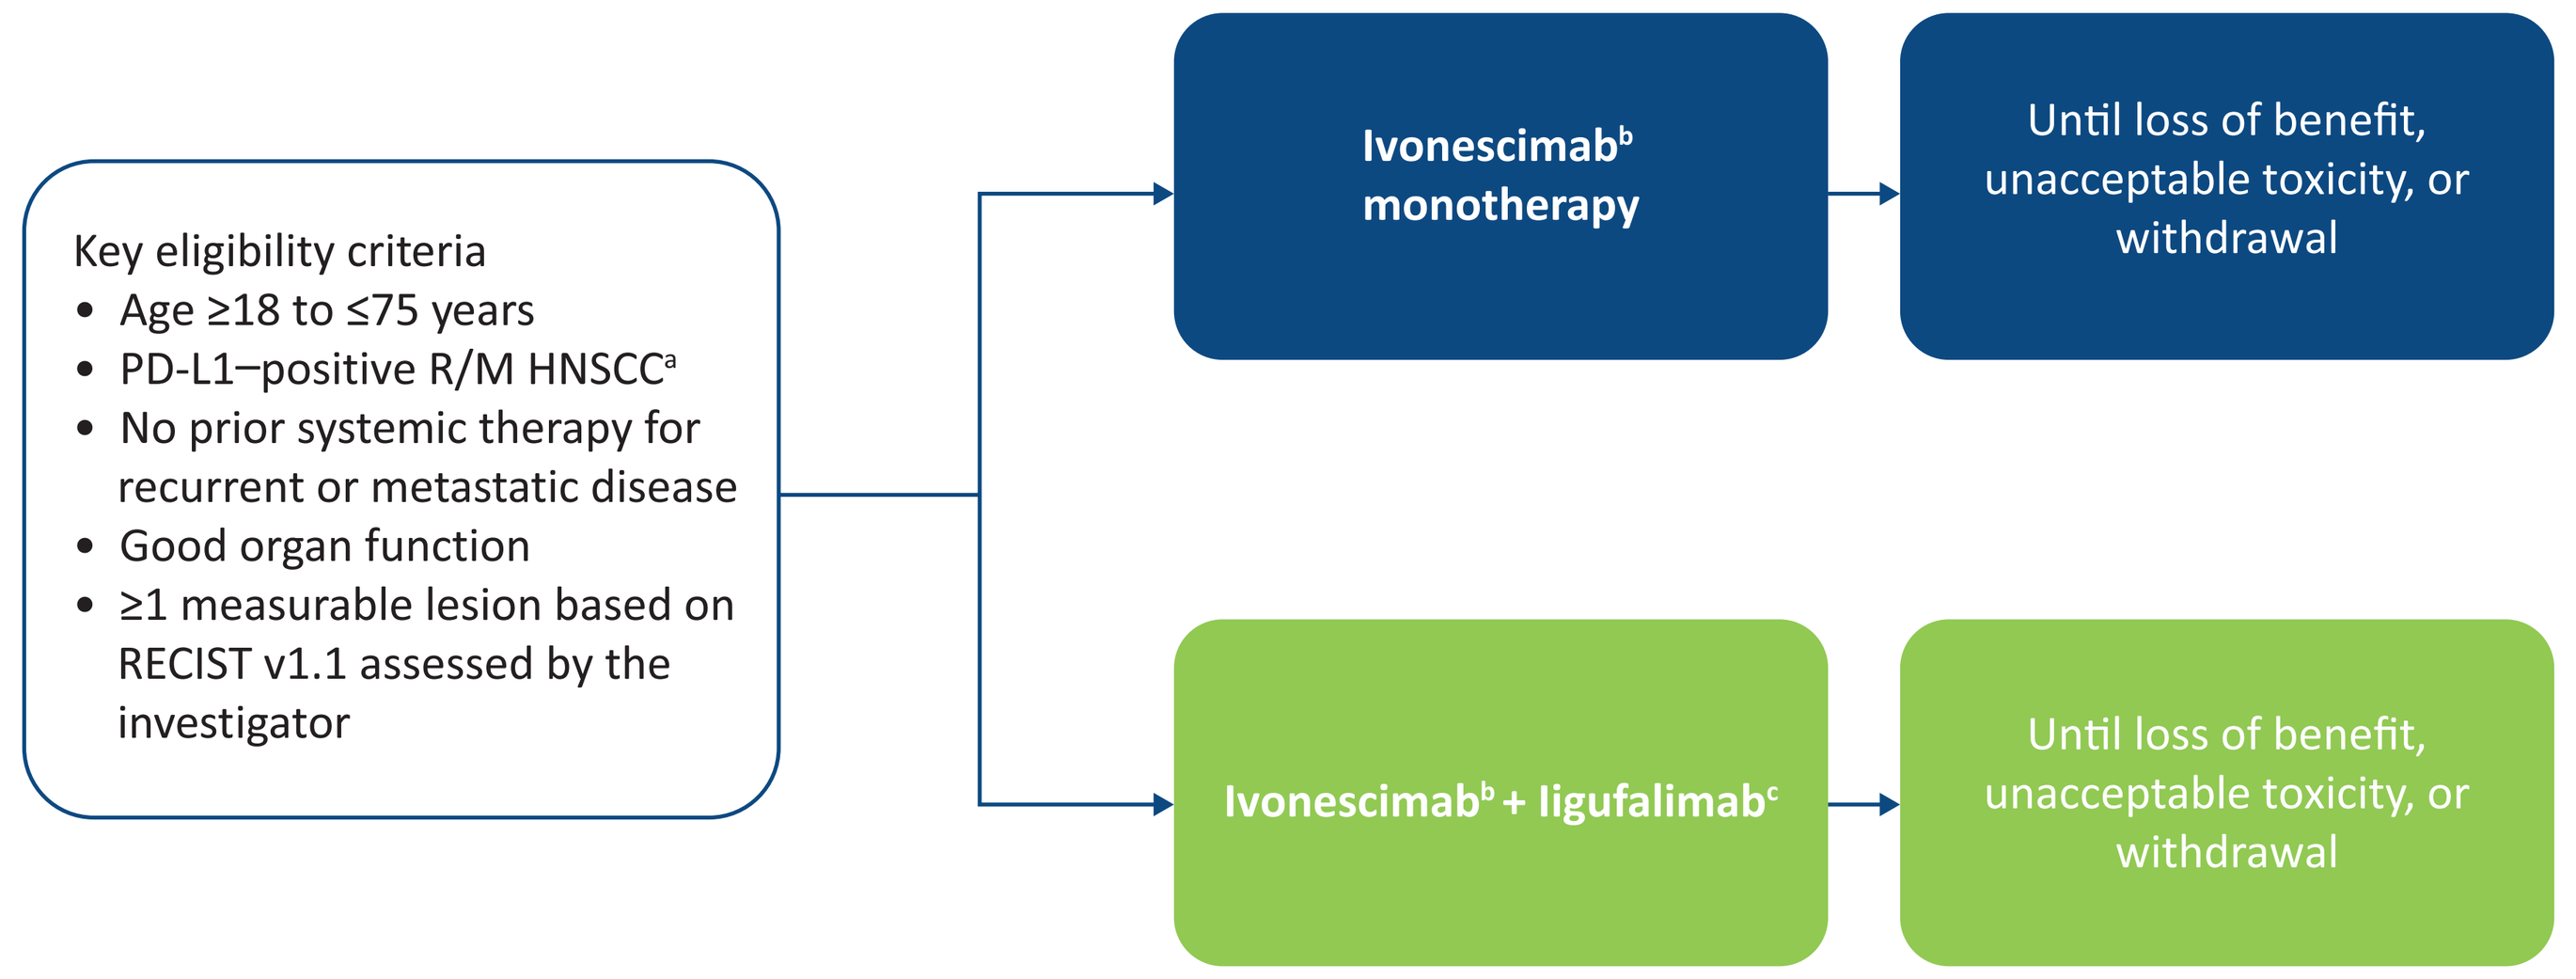

Key eligibility criteria
• Age ≥18 to ≤75 years
• PD-L1–positive R/M HNSCC[a]
• No prior systemic therapy for recurrent or metastatic disease
• Good organ function
• ≥1 measurable lesion based on RECIST v1.1 assessed by the investigator
Ivonescimab[b] monotherapy
Until loss of benefit, unacceptable toxicity, or withdrawal
Ivonescimab[b] + ligufalimab[c]
Until loss of benefit, unacceptable toxicity, or withdrawal

HNSCC, head and neck squamous cell carcinoma; Q3W, once every 3 weeks; PD-L1, programmed death-ligand 1; RECIST v1.1, Response Evaluation Criteria in Solid Tumors, version 1.1; R/M, recurrent/metastatic.

[a]Histologically or cytologically confirmed R/M HNSCC with a PD-L1 combined positive score of ≥1.

[b]Ivonescimab 10 mg/kg Q3W.

[c]Ligufalimab 45 mg/kg Q3W.

Outcomes

- The primary safety end points were incidence and severity of adverse events (AEs), and clinically significant abnormal laboratory findings
- The primary efficacy end point was investigator-assessed objective response rate (ORR) based on Response Evaluation Criteria in Solid Tumors, version 1.1 (RECIST v1.1)
- Key secondary end points were disease control rate (DCR), duration of response (DOR), and progression-free survival (PFS) based on RECIST v1.1

RESULTS

Participants

- As of March 19, 2024, a total of 30 patients (10 in the ivonescimab monotherapy group; 20 in the ivonescimab + ligufalimab group) with R/M HNSCC were enrolled in the study
- Most enrolled patients were male (86.7%), with a median age of 60 years (range, 34-75 years; **Table 1**)
- All enrolled patients (100%) had an Eastern Cooperative Oncology Group performance status score of 1 and 17 (57%) had a PD-L1 combined positive score of ≥20 at baseline
- The median duration of follow-up was 3.3 months (95% CI, 0.9-6.4) for the ivonescimab monotherapy group and 4.1 months (95% CI, 2.8-4.8) for the ivonescimab + ligufalimab group
- The median duration of exposure to study treatment was 1.9 months (range, 0.7-6.4) in the ivonescimab monotherapy group and 3.9 months (range, 1.4-10.1) in the ivonescimab + ligufalimab group

Table 1. Baseline Characteristics

Characteristic	Ivonescimab n = 10	Ivonescimab + ligufalimab n = 20
Age, median (range), years	59.2 (34.3-67.2)	60.8 (41.8-75.2)
Sex, n (%)		
Male	8 (80.0)	18 (90.0)
Female	2 (20.0)	2 (10.0)
ECOG PS score, n (%)		
1	10 (100)	20 (100)
Smoking status, n (%)		
Current	3 (30.0)	2 (10.0)
Former	2 (20.0)	10 (50.0)
Never	5 (50.0)	8 (40.0)
Primary tumor location, n (%)		
Hypopharynx	0	3 (15.0)
Larynx	2 (20.0)	6 (30.0)
Oral cavity	7 (70.0)	4 (20.0)
Oropharynx	1 (10.0)	7 (35.0)
Disease status, n (%)		
Metastatic	2 (20.0)	9 (45.0)
Recurrent only	8 (80.0)	10 (50.0)
Newly diagnosed, nonmetastatic	0	1 (5.0)
PD-L1 CPS, n (%)		
CPS ≥1 and <20	4 (40.0)	9 (45.0)
CPS ≥20	6 (60.0)	11 (55.0)

CPS, combined positive score; ECOG PS, Eastern Cooperative Oncology Group performance status; PD-L1, programmed death-ligand 1.
Race/ethnicity demographics data were not collected.

Safety

- Treatment-related adverse events (TRAEs) were reported in 20 patients (66.6%); 1 was grade ≥3 (3.3%), and none led to discontinuation of treatment or death (**Table 2**)
- The most common TRAEs across both treatment groups were proteinuria (16.7%) and hypothyroidism (13.3%; **Figure 2**)

Table 2. Summary of Safety Results

Adverse event	Ivonescimab n = 10	Ivonescimab + ligufalimab n = 20
TEAEs, n (%)		
Grade ≥3	1 (10.0)	5 (25.0)
Leading to discontinuation	0	1 (5.0)
Serious TEAE	1 (10.0)	4 (20.0)
Leading to death	1 (10.0)	3 (15.0)
TRAEs, n (%)		
Grade ≥3	0	1 (5.0)[a]
Leading to discontinuation	0	0
Serious TRAE	0	1 (5.0)
Leading to death	0	0

TEAE, treatment-emergent adverse event; TRAE, treatment-related adverse event.

[a]1 patient reported grade ≥3 TRAEs in the ivonescimab + ligufalimab group (decreased appetite and tumor hemorrhage).

RESULTS (CONTINUED)

Figure 2. Most Common TRAEs (≥10% overall)[a]

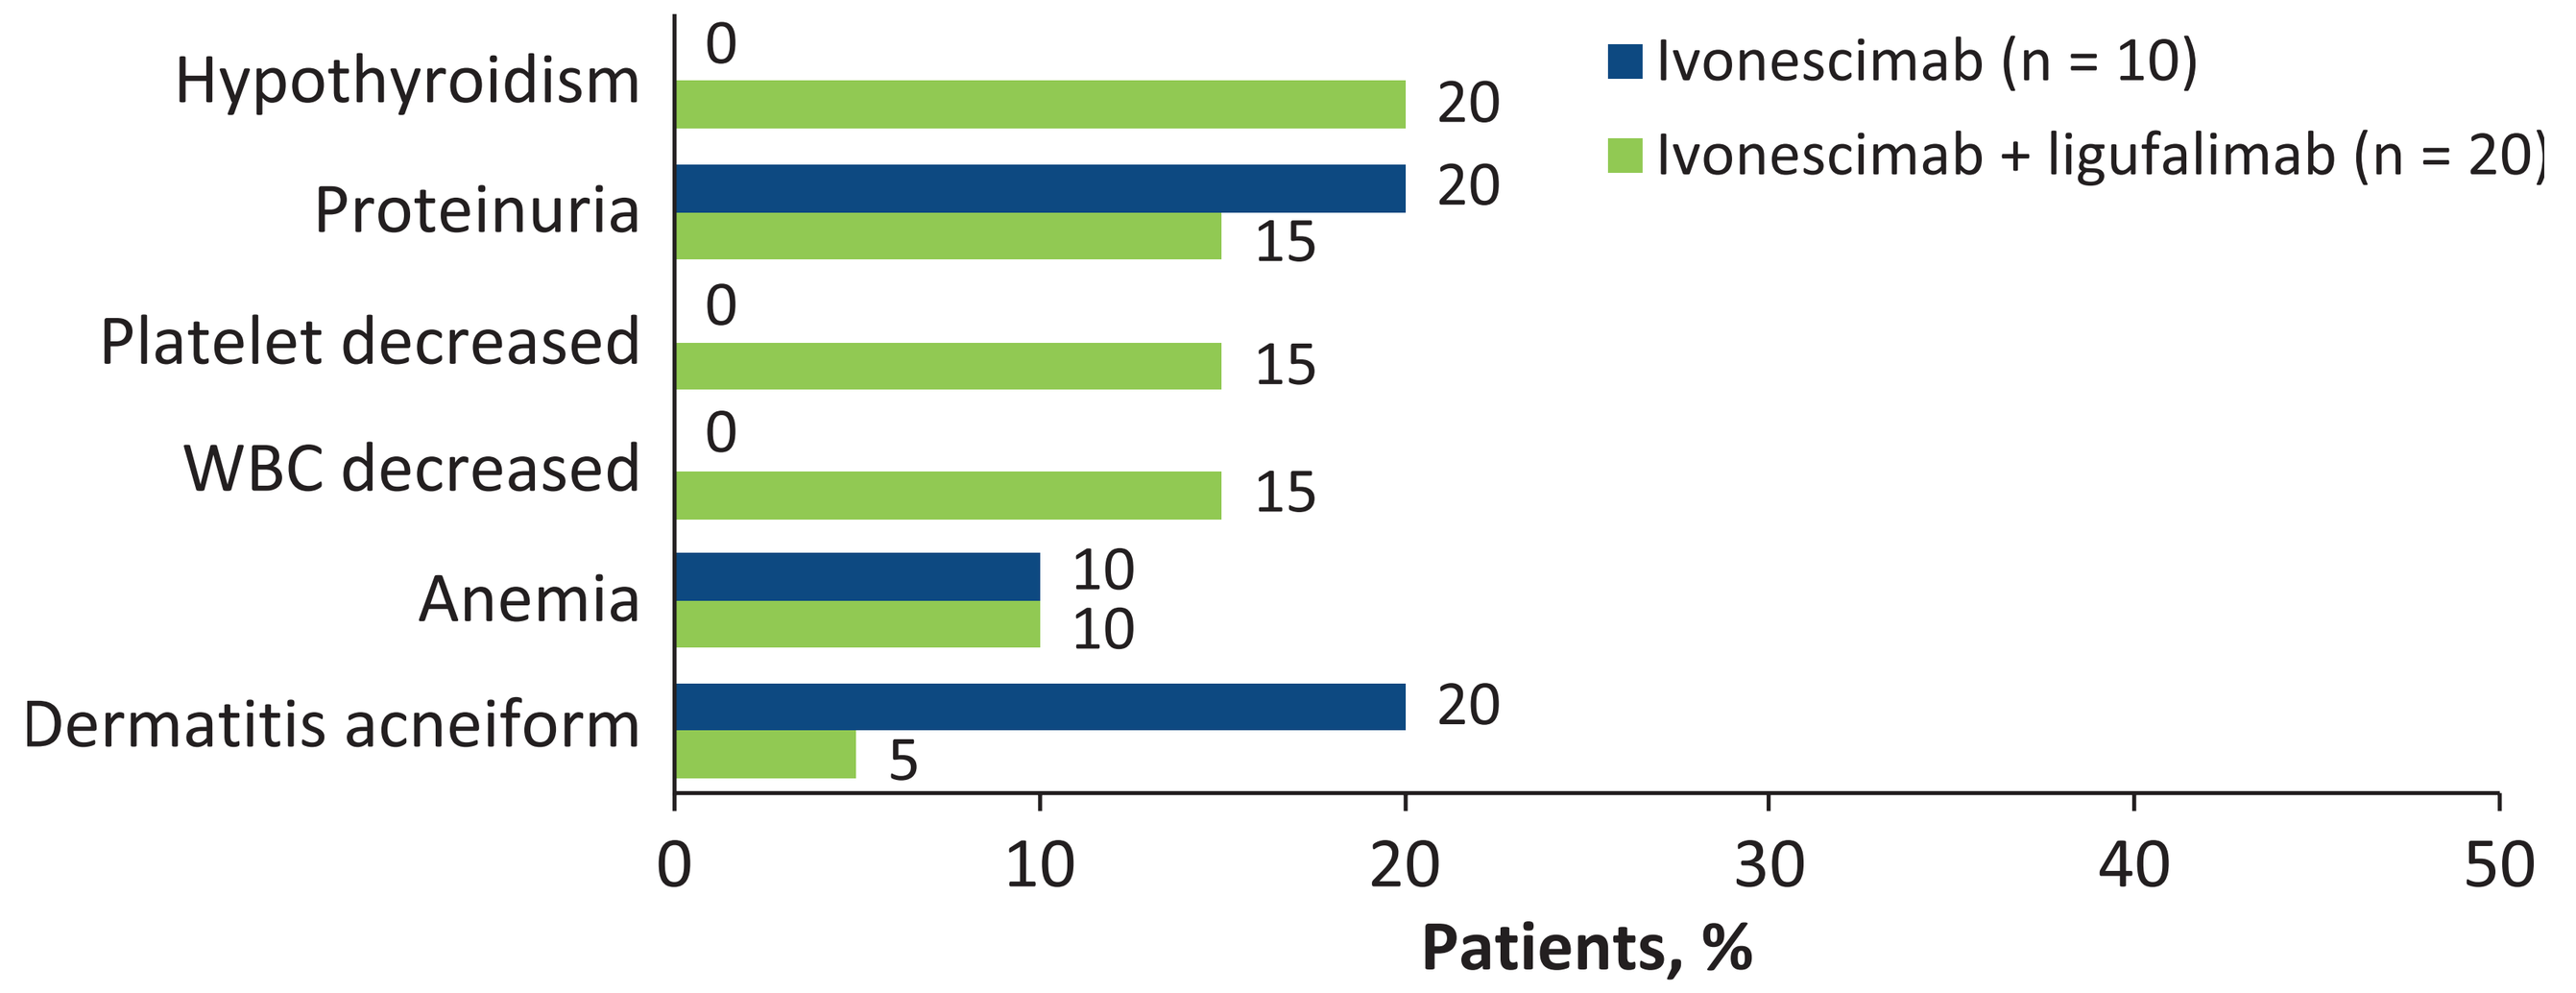

Ivonescimab (n = 10)
Ivonescimab + ligufalimab (n = 20)
Hypothyroidism 0 20
Proteinuria 20 15
Platelet decreased 0 15
WBC decreased 0 15
Anemia 10 10
Dermatitis acneiform 20 5
0 10 20 30 40 50
Patients, %

TRAE, treatment-related adverse event; WBC, white blood cell.

[a]All of the most common TRAEs ≥10% were grades 1 to 2.

Efficacy

- At the time of data cutoff, ORR by investigator assessment was 30% (3/10) in the ivonescimab monotherapy group and 60% (12/20) in the ivonescimab + ligufalimab group (**Table 3**)
- DCR was 80% (8/10) in the ivonescimab monotherapy group and 90% (18/20) in the ivonescimab + ligufalimab group
- Median DOR was not reached in either treatment group
- Individual patient-level responses at the time of data cutoff are shown in **Figures 3 and 4**

Table 3. Overview of Objective Response Rate

Response	Ivonescimab n = 10	Ivonescimab + ligufalimab n = 20
ORR %, (95% CI)		
Overall	30.0 (6.7-65.2)[a]	60.0 (36.1-80.9)[a]
PD-L1 CPS ≥1 and <20	75.0 (19.4-99.4)[a]	44.4 (13.7-78.8)[a]
PD-L1 CPS ≥20	0	72.7 (39.0-94.0)
BOR, n (%)		
CR	0	1 (5.0)
PR	3 (30.0)	11 (55.0)
SD	5 (50.0)	6 (30.0)
PD	2 (20.0)	2 (10.0)

BOR, best overall response; CPS, combined positive score; CR, complete response; ORR, objective response rate; PD, progressive disease; PD-L1, programmed death-ligand 1; PR, partial response; SD, stable disease.

[a]2 patients (n = 1 in each treatment group) with SD subsequently had PR after data cutoff. This resulted in an ORR of 40.0% and 65.0%, respectively, in the ivonescimab group and ivonescimab + ligufalimab groups overall and 80.0% and 55.5% in the PD-L1 CPS≥1 and <20 subgroup.

Figure 3. Best Percentage Change From Baseline in Sum of Tumor Diameters

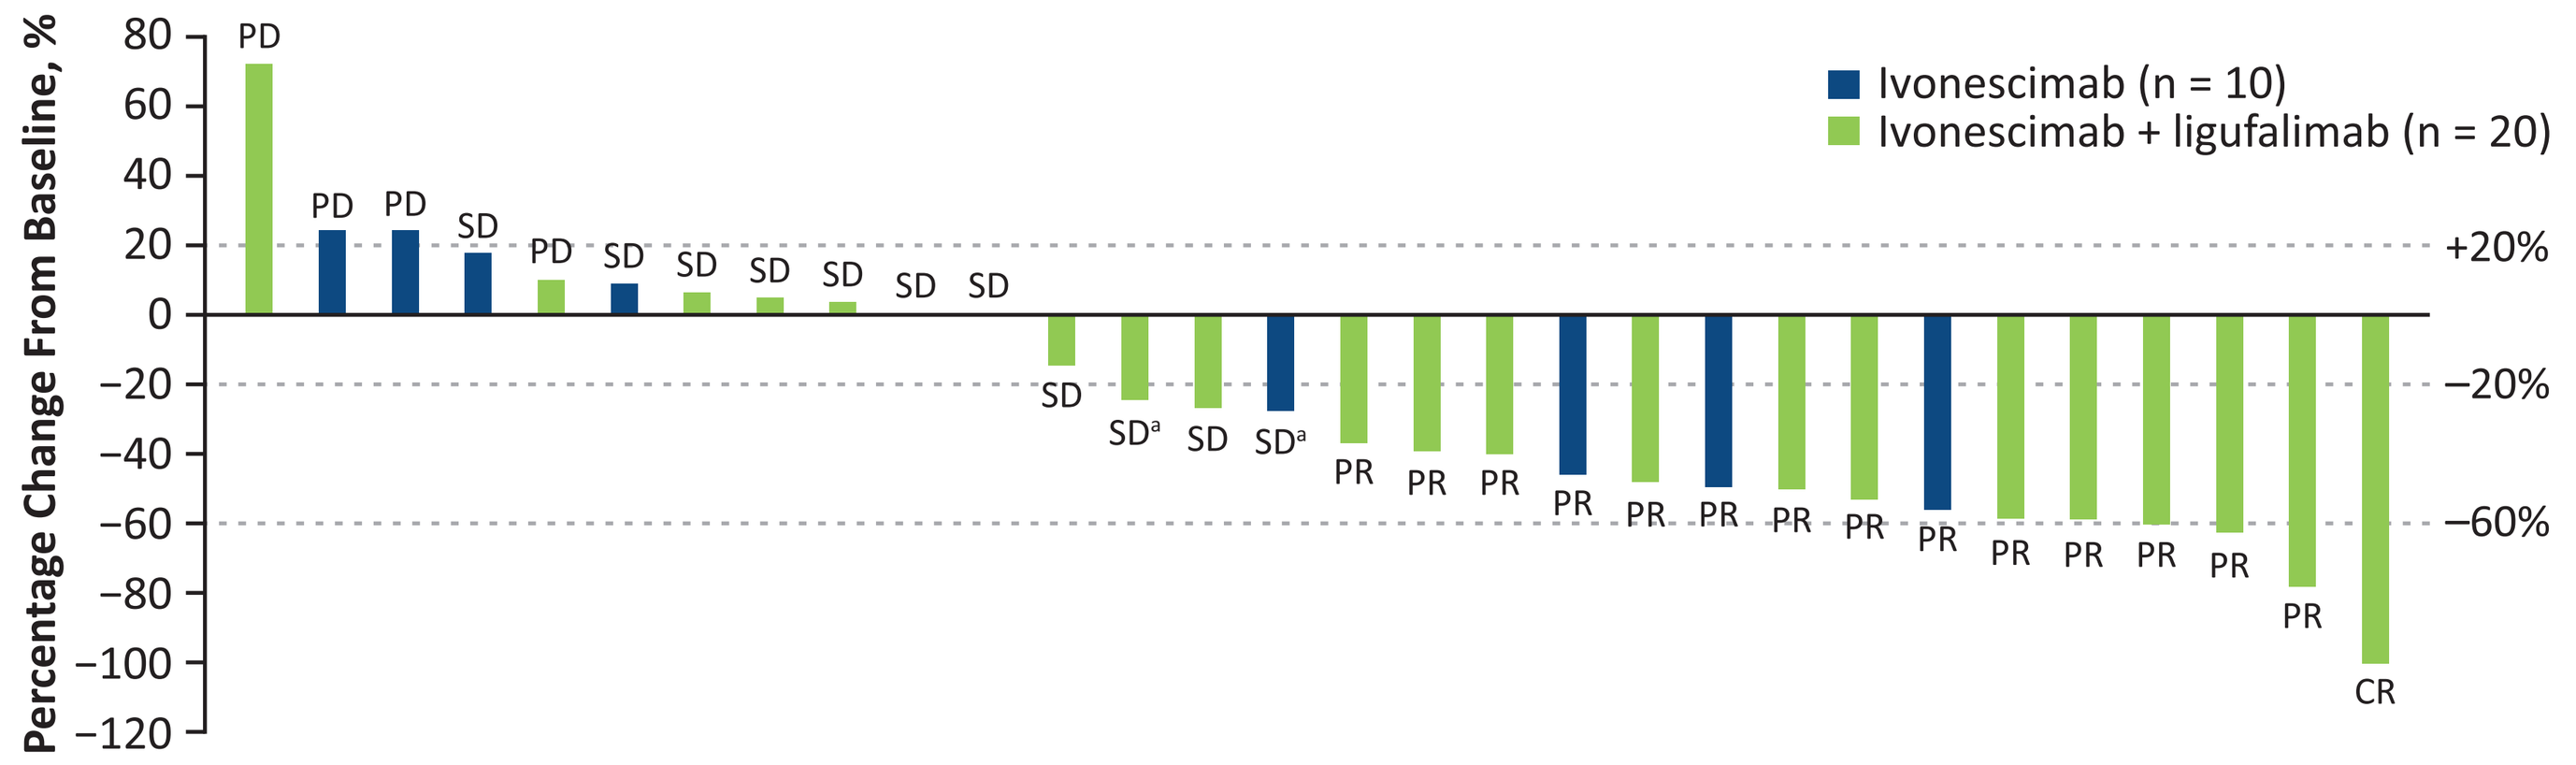

Ivonescimab (n = 10)
Ivonescimab + ligufalimab (n = 20)
Percentage Change From Baseline, %
+20%
−20%
−60%
PD PD PD SD PD SD SD SD SD SD SD
SD SD[a] SD SD[a] PR PR PR PR PR PR PR PR PR PR PR PR PR PR PR CR

CR, complete response; PD, progressive disease; PR, partial response; SD, stable disease.

[a]Patient subsequently had PR after data cutoff.

Figure 4. Percentage Change From Baseline in Sum of Diameters Over Time

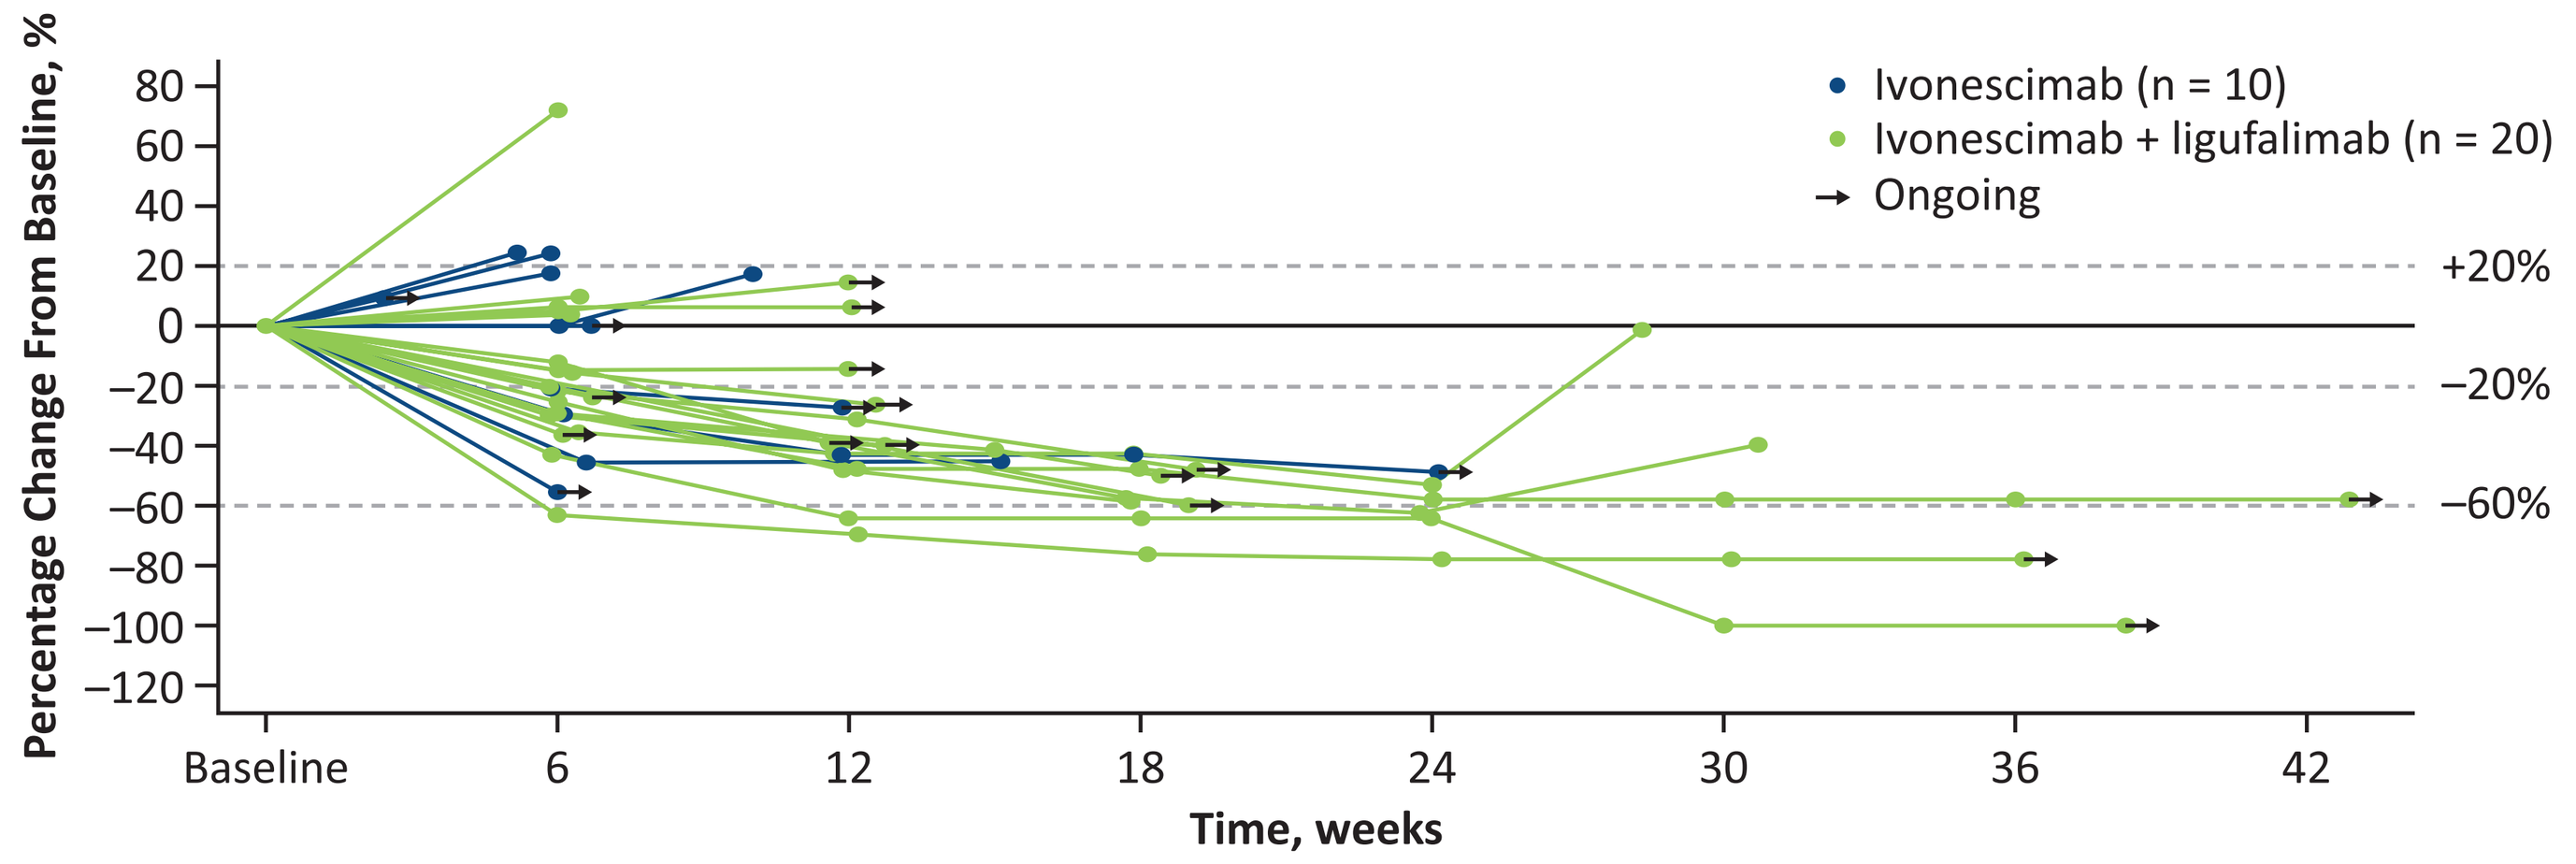

Ivonescimab (n = 10)
Ivonescimab + ligufalimab (n = 20)
Ongoing
Percentage Change From Baseline, %
+20%
−20%
−60%
Baseline 6 12 18 24 30 36 42
Time, weeks

RESULTS (CONTINUED)

- Median PFS was 5.0 months (95% CI, 1.2-NE) for the ivonescimab monotherapy group; the 6-month PFS rate was not reached
- Median PFS in the ivonescimab + ligufalimab group was 7.1 months (95% CI, 3.6-NE), with a 6-month PFS rate of 71.8% (**Figure 5**)

Figure 5. Kaplan Meier Curve of Progression-Free Survival

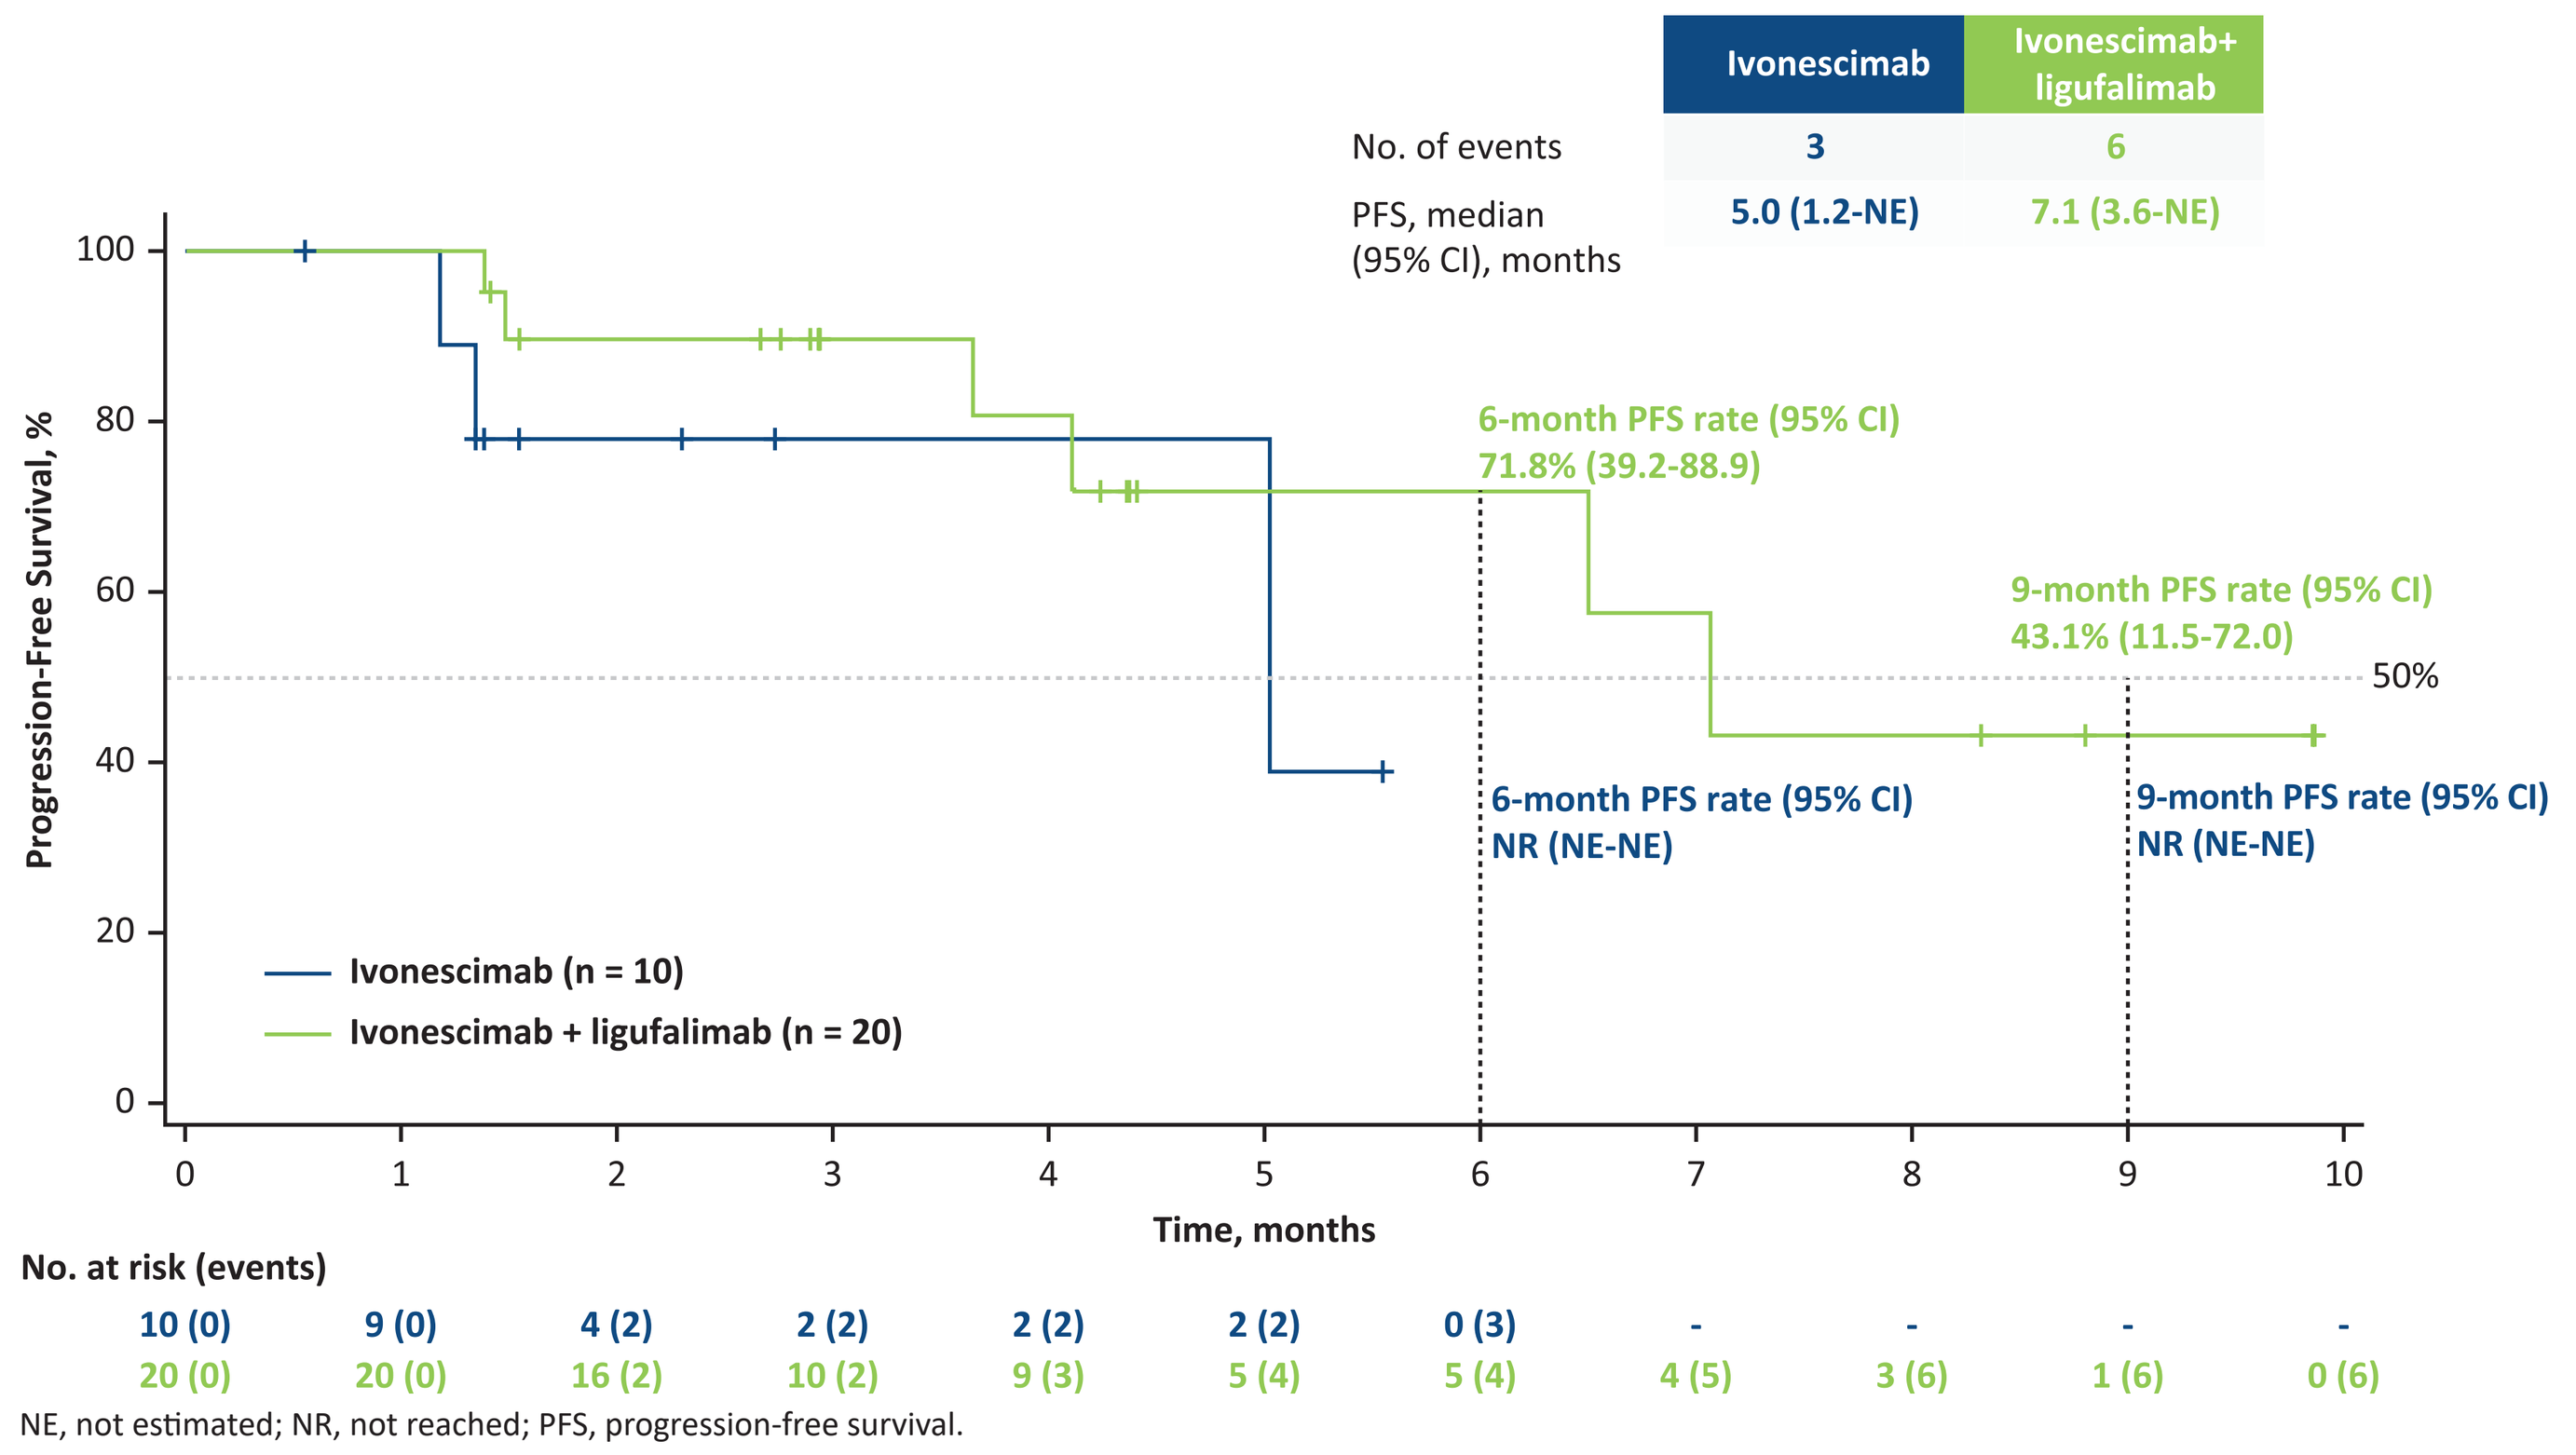

Ivonescimab | Ivonescimab+ ligufalimab
No. of events 3 6
PFS, median (95% CI), months 5.0 (1.2-NE) 7.1 (3.6-NE)
Progression-Free Survival, %
6-month PFS rate (95% CI) 71.8% (39.2-88.9)
9-month PFS rate (95% CI) 43.1% (11.5-72.0)
50%
6-month PFS rate (95% CI) NR (NE-NE)
9-month PFS rate (95% CI) NR (NE-NE)
Ivonescimab (n = 10)
Ivonescimab + ligufalimab (n = 20)
Time, months
No. at risk (events)
10 (0) 9 (0) 4 (2) 2 (2) 2 (2) 2 (2) 0 (3) - - - -
20 (0) 20 (0) 16 (2) 10 (2) 9 (3) 5 (4) 5 (4) 4 (5) 3 (6) 1 (6) 0 (6)

NE, not estimated; NR, not reached; PFS, progression-free survival.

CONCLUSIONS

- Ivonescimab as monotherapy or in combination with ligufalimab showed promising antitumor activity and an acceptable safety profile in patients with PD-L1–positive R/M HNSCC
- Results of this analysis support further evaluation of ivonescimab as monotherapy or in combination with ligufalimab in the treatment of patients with R/M HNSCC

ACKNOWLEDGMENTS

The authors thank the participants and their caregivers, the investigators, and all the site staff who have been participating in this study. This study is sponsored by Akeso Biopharma, Inc. Medical writing assistance was provided by ApotheCom (Yardley, PA, USA) and was funded by Summit Therapeutics.

REFERENCES

1. Machiels JP et al. *Ann Oncol*. 2020;31:1462-1475.
2. Bai S et al. *Transl Lung Cancer Res*. 2021;10:2614-2624.
3. Wu L et al. *Oncoimmunology*. 2018;7:e1397248.
4. Liu Y et al. *J Adv Res*. 2024;63:129-158.
5. Zhao Y et al. *EClinicalMedicine*. 2023;62:102106.
6. Wang L et al. *J Thorac Oncol*. 2024;19:465-474.
7. Frentzas S et al. *J Immunother Cancer*. 2024;12:e008037.
8. Qu T et al. *J Immunother Cancer*. 2022;10:e005517.
9. Gan HK et al. *J Clin Oncol*. 2021;39(15 suppl):2630.

DISCLOSURES

W.L., Z.M.W., B.L., and M.X. are employees of Akeso Biopharma, Inc. All other authors have no conflicts of interest to disclose.

CONTACT INFORMATION

Please contact the presenting author, Xiaozhong Chen, at cxzfyun@sina.com for questions or comments.

Copies of this poster obtained through QR (Quick Response) and/or text key codes are for personal use only and may not be reproduced without written permission from of the authors.

By use of the QR Code, you agree to Summit's privacy notice, located at smmttx.com.

Presented at the European Society for Medical Oncology (ESMO) Congress 2024; September 13-17, 2024; Barcelona, Spain